Additional Efficacy Analyses From the Phase 2b SYNERGY Study Investigating the Anti-LINGO-1 Monoclonal Antibody Opicinumab for Treatment of Disabling Multiple Sclerosis

P5.337

Hupperts R,[1] Mellion M,[2,*] Edwards KR,[3] Drulović J,[4] Montalban X,[5] Hartung H-P,[6] Brochet B,[7] Calabresi PA,[8] Rudick R,[2] Ibrahim A,[9] Zhang Y,[2] Cadavid D,[10,*] Xu L,[2] on behalf of the SYNERGY investigators

[1]Orbis Medisch Centrum, Maastricht University Medical Center, Sittard, The Netherlands; [2]Biogen, Cambridge, MA, USA; [3]Multiple Sclerosis Center of Northeastern New York, Latham, NY, USA; [4]Clinic of Neurology, CCS, Faculty of Medicine, University of Belgrade, Belgrade, Serbia; [5]Vall d'Hebron University Hospital, Barcelona, Spain; [6]Department of Neurology, Heinrich-Heine University Düsseldorf, Düsseldorf, Germany; [7]University Hospital of Bordeaux and University of Bordeaux, Bordeaux, France; [8]The Johns Hopkins Multiple Sclerosis Center, Baltimore, MD, USA; [9]Biogen, Maidenhead, UK; [10]Fulcrum Therapeutics, Cambridge, MA, USA

*Former employee of Biogen

Conclusions

- The SYNERGY primary endpoint analyses showed an unexpected inverted U-shaped dose response.
 - The hypothesis of a linear trend dose response to opicinumab was rejected.
- Additional prespecified analyses evaluating improvement on components of the multicomponent primary endpoint revealed that 10 mg/kg opicinumab demonstrated a consistent treatment response vs. placebo.
 - The estimated proportion of participants with improvement on the alternative multicomponent endpoint was highest for 10 mg/kg opicinumab compared with placebo and the other opicinumab doses.
 - Similarly, the hazard ratio for time to improvement was highest for 10 mg/kg opicinumab vs. the other treatment arms.
- Additional study, particularly of the 10-mg/kg dose, is necessary to further characterize the potential treatment effects of opicinumab in patients with pre-existing disability.
- More details on the SYNERGY study are provided in posters P2.107, P5.368, and P6.332 and oral presentation S33.004.

Introduction

- Opicinumab (anti-LINGO-1) is a novel candidate neuroreparative agent previously assessed in the RENEW (Phase 2a; NCT01721161) trial of participants with a first episode of optic neuritis.[1]
- SYNERGY (Phase 2b; NCT01864148) was designed to assess the efficacy and safety of escalating doses of opicinumab vs. placebo in participants with relapsing multiple sclerosis (MS; relapsing-remitting MS [RRMS], 80%; secondary progressive MS [SPMS], 20%) when used for 72 weeks concurrently with intramuscular (IM) interferon (IFN) beta-1a.
 - Doses up to 100 mg/kg were tested as opicinumab is a monoclonal antibody with low blood-brain barrier penetration (cerebrospinal fluid:serum ratio, ~0.1%).[2]
- Additional prespecified analyses of improvement in pre-existing disability (including using an alternative endpoint minus the 3-Second Paced Auditory Serial Addition Test [PASAT-3]) may provide further evidence for the potential benefit of opicinumab in patients with relapsing MS.

Objectives

- To further investigate the potential benefit of opicinumab in SYNERGY participants using prespecified analyses of neurofunctional improvement in pre-existing disability.

Methods

- SYNERGY was a randomized placebo-controlled dose-ranging study (Figure 1).
- Participants were 18–58 years of age, had active RRMS or relapsing SPMS, and an Expanded Disability Status Scale (EDSS) score of 2.0–6.0.
- The primary endpoint was the percentage of participants with improvement in disability and neurophysical and/or cognitive function over 72 weeks measured by a multicomponent endpoint comprising: EDSS, Timed 25-Foot Walk (T25FW), 9-Hole Peg Test (9HPT; dominant and nondominant hand), and PASAT-3.
 - Improvement was defined as: ≥1.0-point decrease in EDSS score (from a baseline score ≤6.0) and/or ≥15% improvement from baseline in T25FW, 9HPT, or PASAT-3, all confirmed for ≥3 months. Improvement in any of the measures needed to be confirmed in the same measure.
- Prespecified additional analyses included confirmed improvement and time to confirmed improvement on:
 - An alternative multicomponent improvement endpoint comprising physical components of the primary endpoint: EDSS, T25FW, and 9HPT (dominant and nondominant hand).
 - This was prespecified to remove PASAT-3 as a previous analysis of baseline scores showed minimal if any impairment by MS.[3]
 - Individual components of the primary endpoint.
 - Two or more of the individual physical components of the primary endpoint.
 - To capture individual improvement across >1 component.
- A logistic regression model was used to analyze improvement probability. Kaplan-Meier curves were used to display distribution of time to improvement and Cox proportional hazard models were used to analyze time to improvement.

Figure 1. SYNERGY study design

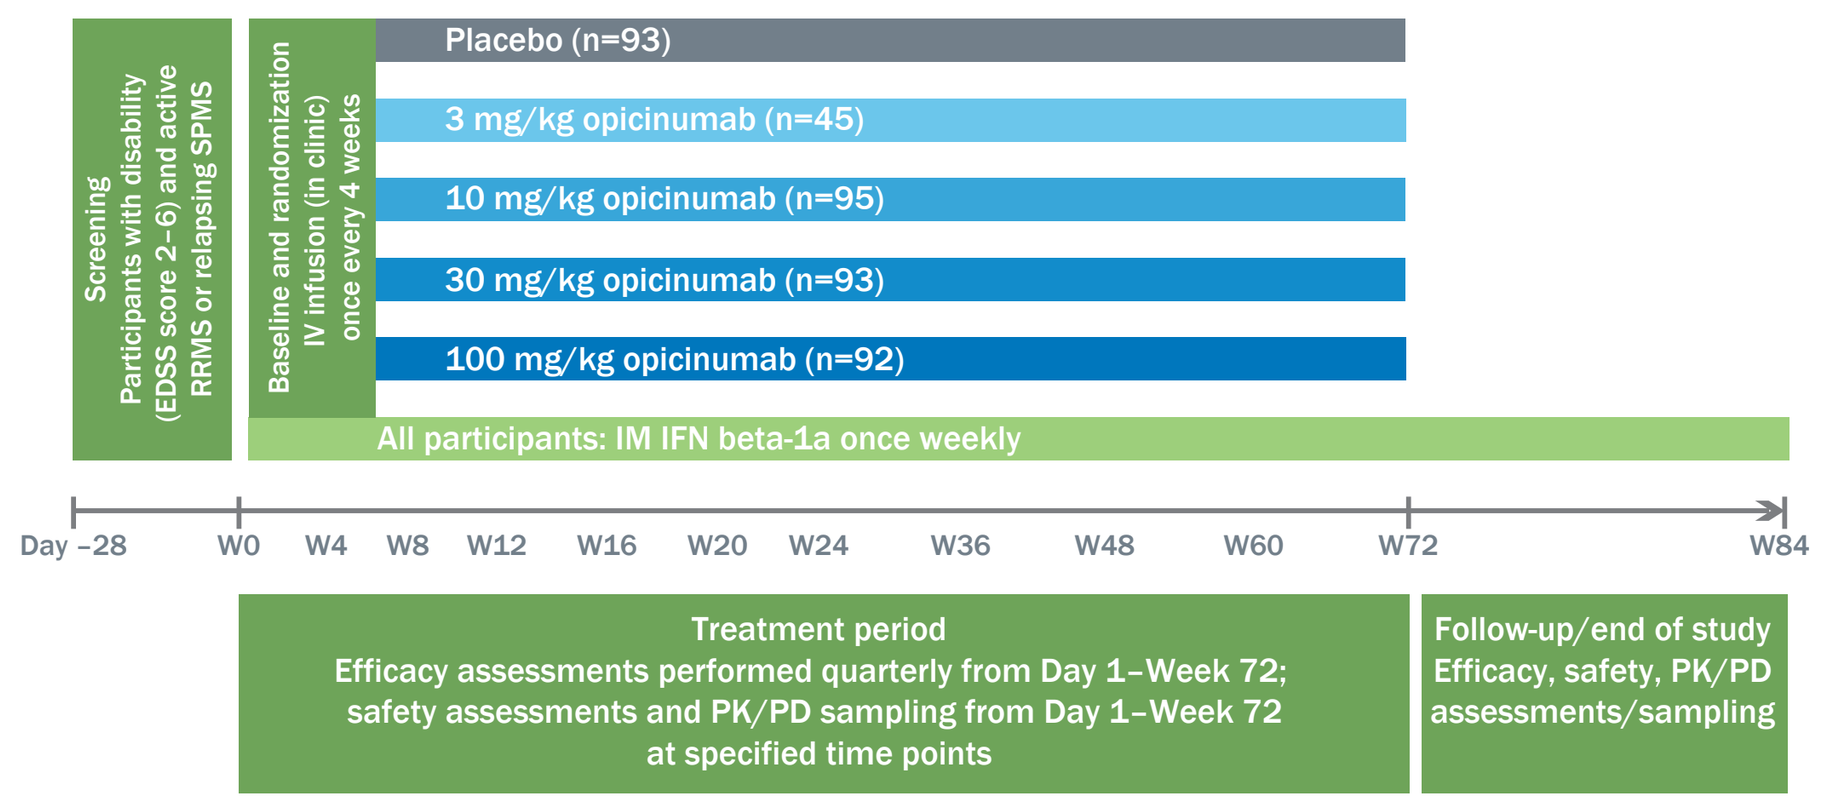
Screening
Participants with disability (EDSS score 2–6) and active RRMS or relapsing SPMS
Baseline and randomization
IV infusion (in clinic) once every 4 weeks
Placebo (n=93)
3 mg/kg opicinumab (n=45)
10 mg/kg opicinumab (n=95)
30 mg/kg opicinumab (n=93)
100 mg/kg opicinumab (n=92)
All participants: IM IFN beta-1a once weekly
Day −28
W0
W4
W8
W12
W16
W20
W24
W36
W48
W60
W72
W84
Treatment period
Efficacy assessments performed quarterly from Day 1–Week 72; safety assessments and PK/PD sampling from Day 1–Week 72 at specified time points
Follow-up/end of study
Efficacy, safety, PK/PD assessments/sampling

IV = intravenous; PD = pharmacodynamics; PK = pharmacokinetics

Results

- Four hundred eighteen participants were randomized and dosed; efficacy was assessed in 412 participants as 6 were excluded from the efficacy analyses due to good clinical practice violations.
- The SYNERGY prespecified primary endpoint using a linear trend test across 5 arms was not met due to a dose response demonstrating an inverted U shape (see oral presentation S33.004).
- The estimated proportion of participants with improvement on the alternative multicomponent improvement endpoint is shown in Figure 2.
 - Analysis of time to improvement on the endpoint is shown in Figure 3.
 - At the dose of 10 mg/kg, opicinumab showed the most favorable odds ratios (ORs) and hazard ratios (HRs) compared with placebo, with a similarly inverted U-shaped dose response.
- Confirmed improvement on individual components of the primary endpoint are shown in Figure 4.
 - For most components, the inverted U-shaped dose response was observed, favoring 10 mg/kg opicinumab.
- Improvement based on ≥2 components of the primary endpoint (EDSS, T25FW, 9HPT, PASAT-3) was 24.5% for placebo and 29.3% (OR, 1.28 [95% CI, 0.51–3.18]), 28.7% (1.24 [0.59–2.58]), 13.9% (0.50 [0.21–1.17]), and 17.2% (0.64 [0.28–1.43]) for 3, 10, 30, and 100 mg/kg opicinumab, respectively.
 - The HRs (95% CIs) for time to improvement on ≥2 components were 1.24 (0.56–2.72), 1.05 (0.56–1.97), 0.53 (0.24–1.16), and 0.74 (0.36–1.53) for 3, 10, 30, and 100 mg/kg opicinumab, respectively, compared with placebo.

Figure 2. Proportion of participants with improvement on the alternative multicomponent endpoint (EDSS, T25FW, 9HPT)

	Placebo n=91	3 mg/kg opicinumab n=45	10 mg/kg opicinumab n=94	30 mgkg opicinumab n=91	100 mg/kg opicinumab n=91
Improvement responders over 72 weeks, n (%)	33 (36)	18 (40)	50 (53)	36 (40)	28 (31)
Estimated proportion of responders[a]	0.416	0.422	0.559	0.447	0.340

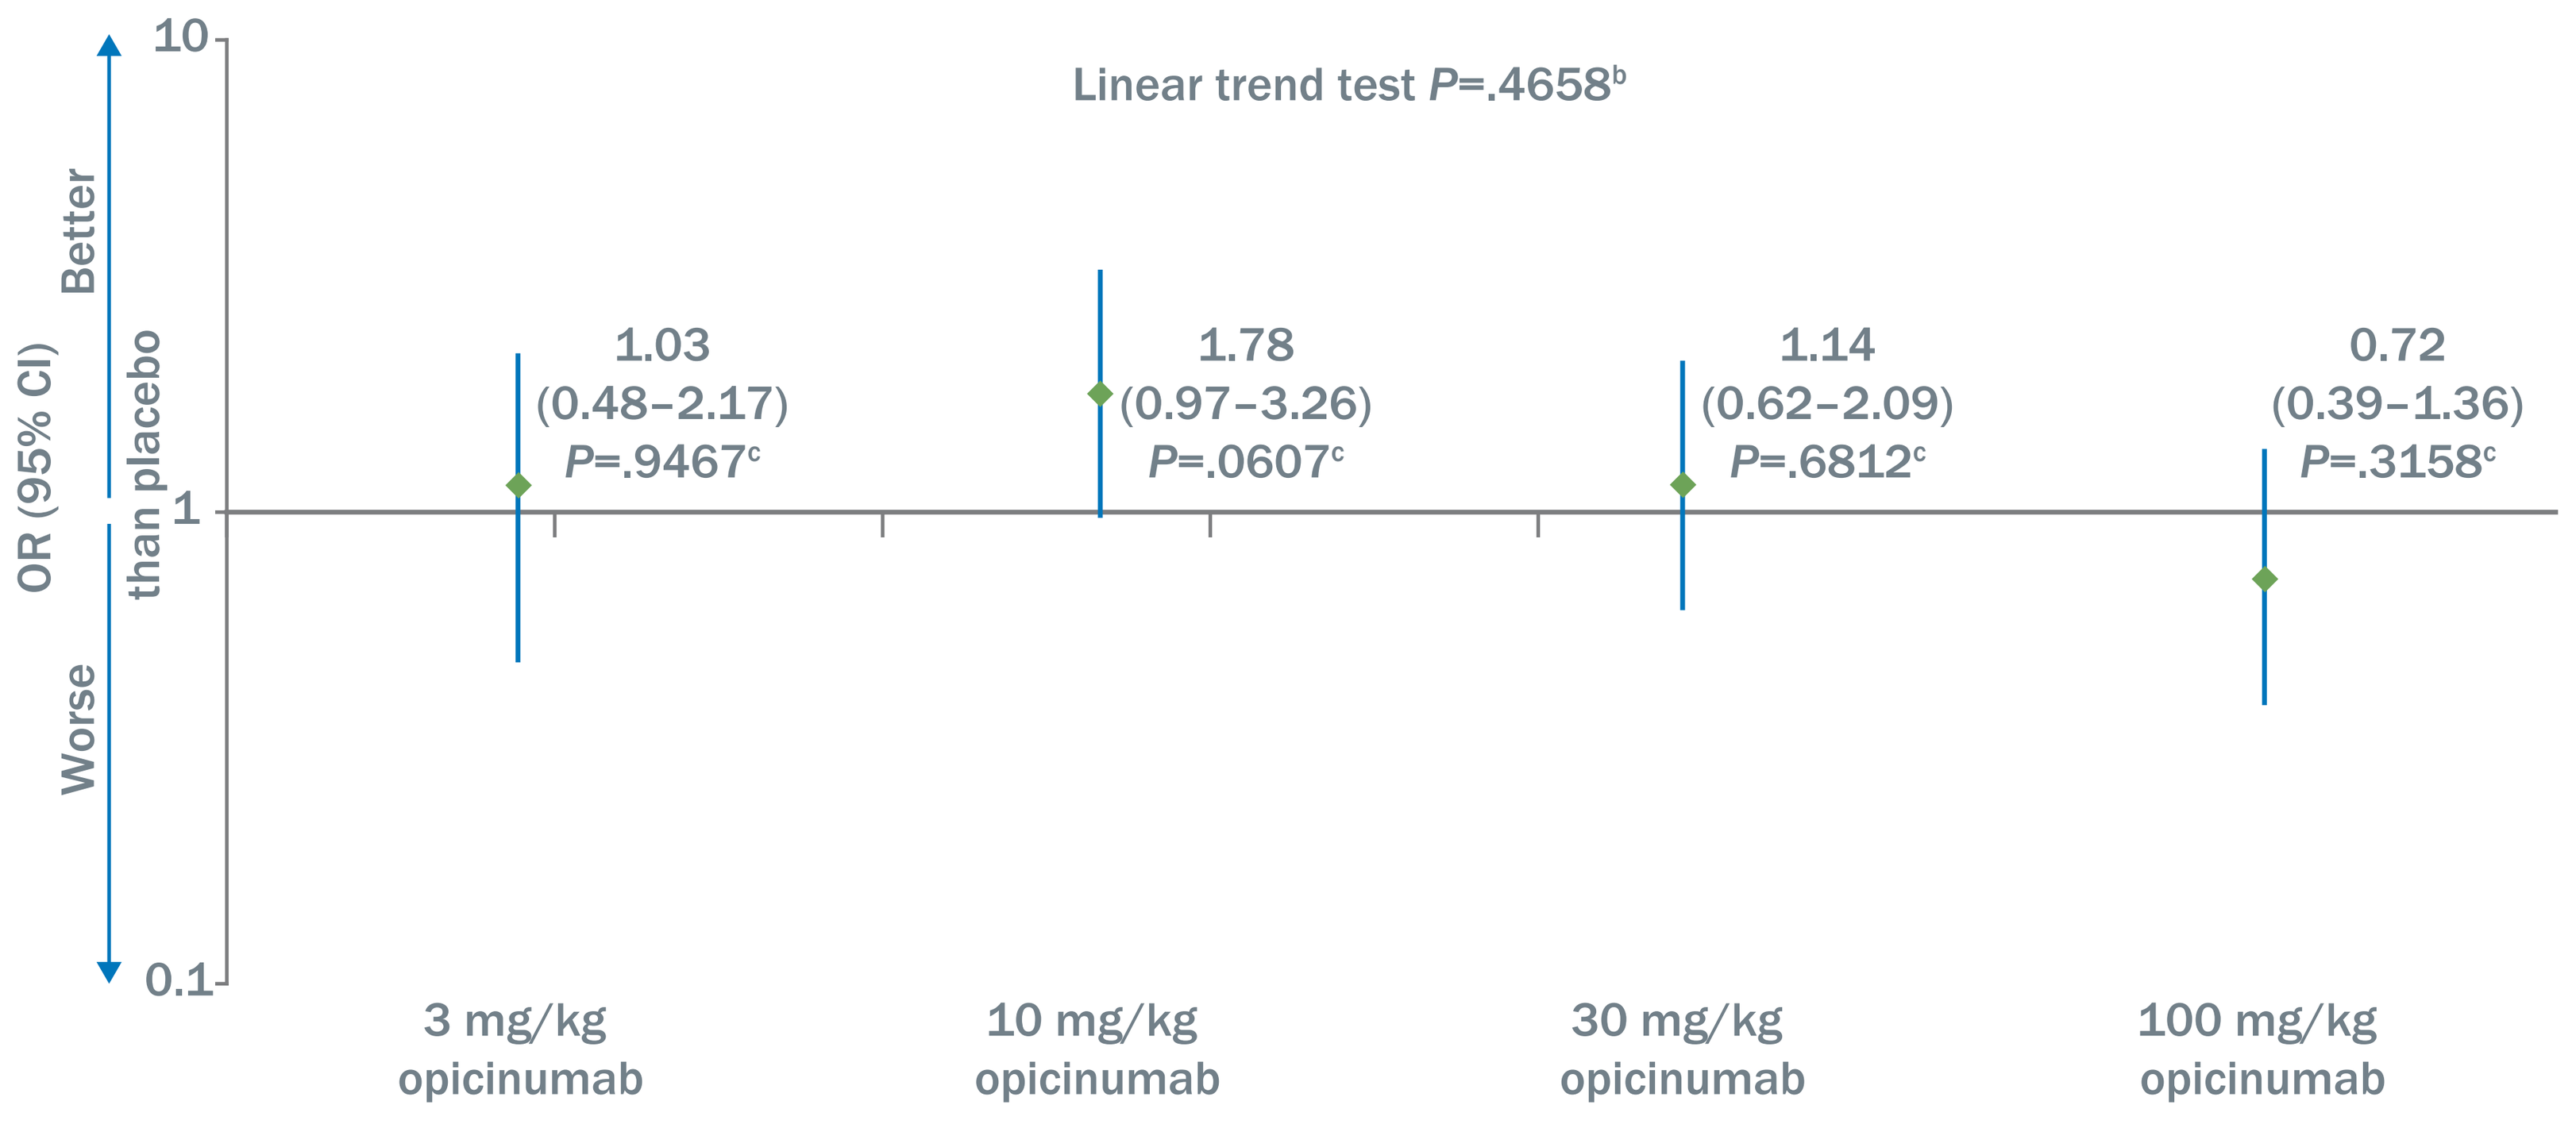
Linear trend test P=.4658[b]
OR (95% CI)
Better than placebo
Worse
10
1
0.1
1.03
(0.48–2.17)
P=.9467[c]
1.78
(0.97–3.26)
P=.0607[c]
1.14
(0.62–2.09)
P=.6812[c]
0.72
(0.39–1.36)
P=.3158[c]
3 mg/kg opicinumab
10 mg/kg opicinumab
30 mg/kg opicinumab
100 mg/kg opicinumab

[a]Based on a logistic regression model adjusted for MS type, region, and baseline component assessments
[b]Based on linear contrast in a logistic regression model
[c]Nominal *P* values for pairwise comparison of each opicinumab arm vs. placebo

Figure 3. Time to improvement on the alternative multicomponent improvement endpoint

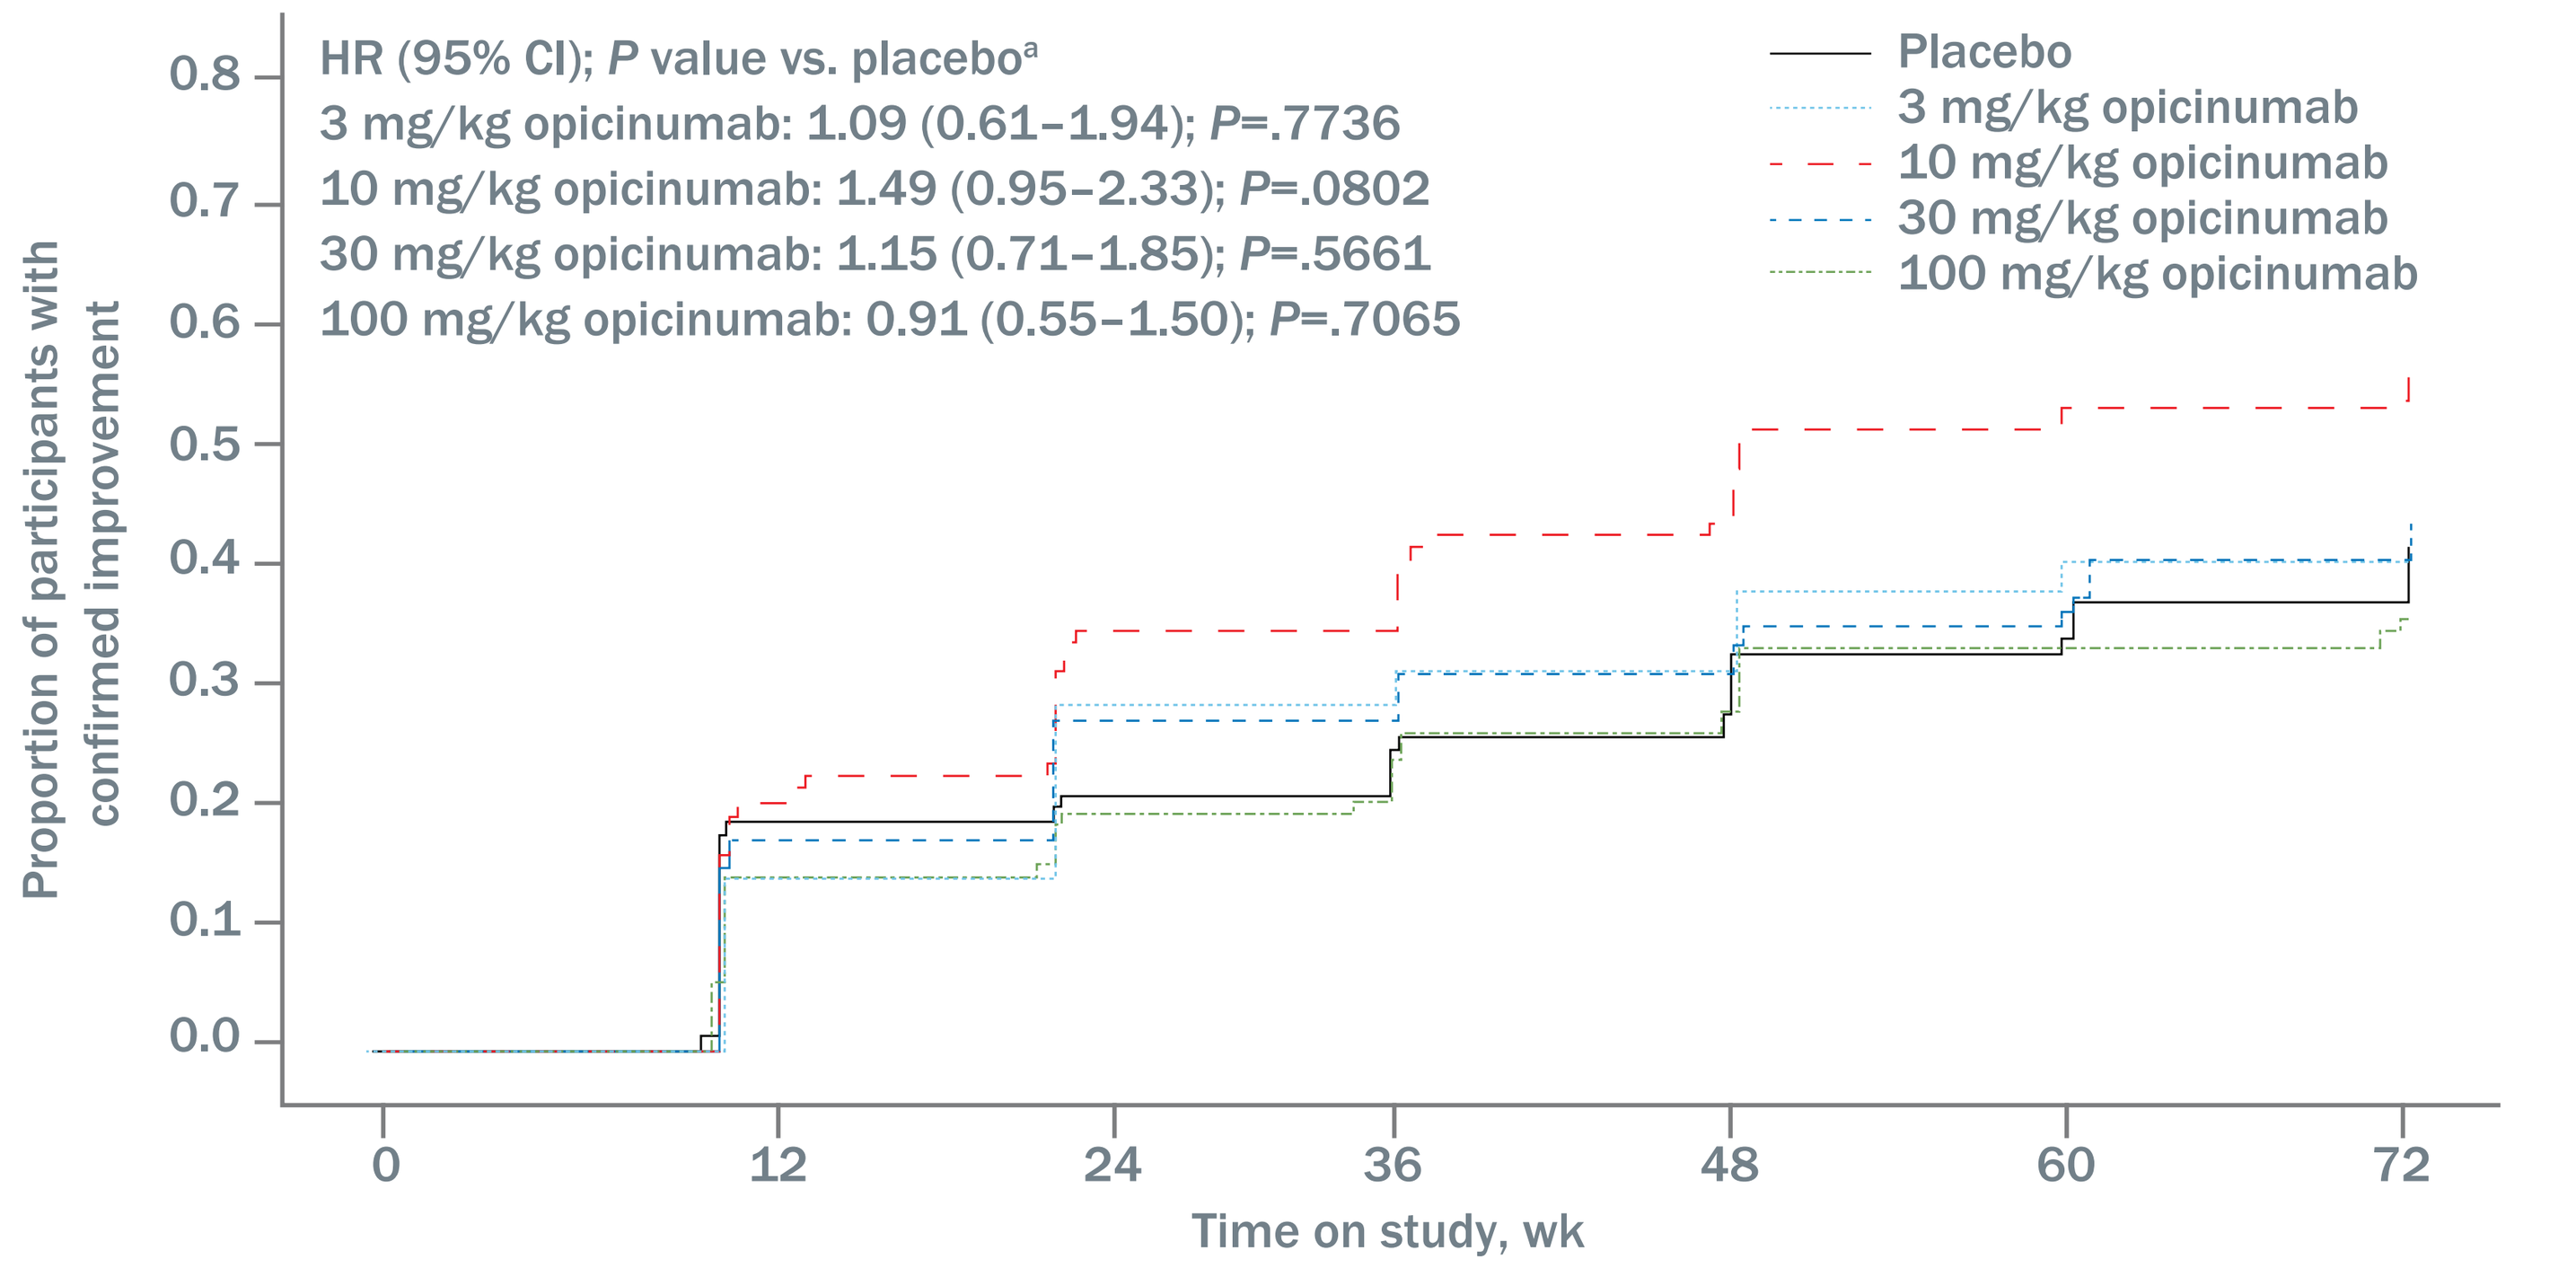
HR (95% CI); P value vs. placebo[a]
3 mg/kg opicinumab: 1.09 (0.61–1.94); P=.7736
10 mg/kg opicinumab: 1.49 (0.95–2.33); P=.0802
30 mg/kg opicinumab: 1.15 (0.71–1.85); P=.5661
100 mg/kg opicinumab: 0.91 (0.55–1.50); P=.7065
Placebo
3 mg/kg opicinumab
10 mg/kg opicinumab
30 mg/kg opicinumab
100 mg/kg opicinumab
Proportion of participants with confirmed improvement
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0.0
0
12
24
36
48
60
72
Time on study, wk

No. of participants at risk							
Placebo	91	82	67	64	55	47	32
3 mg/kg opicinumab	45	41	35	29	29	26	18
10 mg/kg opicinumab	94	91	70	59	50	41	27
30 mg/kg opicinumab	91	82	65	57	51	47	32
100 mg/kg opicinumab	91	74	63	58	49	46	35

[a]Based on a Cox proportional hazards model adjusted for MS type, region, and baseline component assessments

Figure 4. Confirmed improvement responders on individual components of the primary endpoint (EDSS, T25FW, 9HPT, PASAT-3)[a]

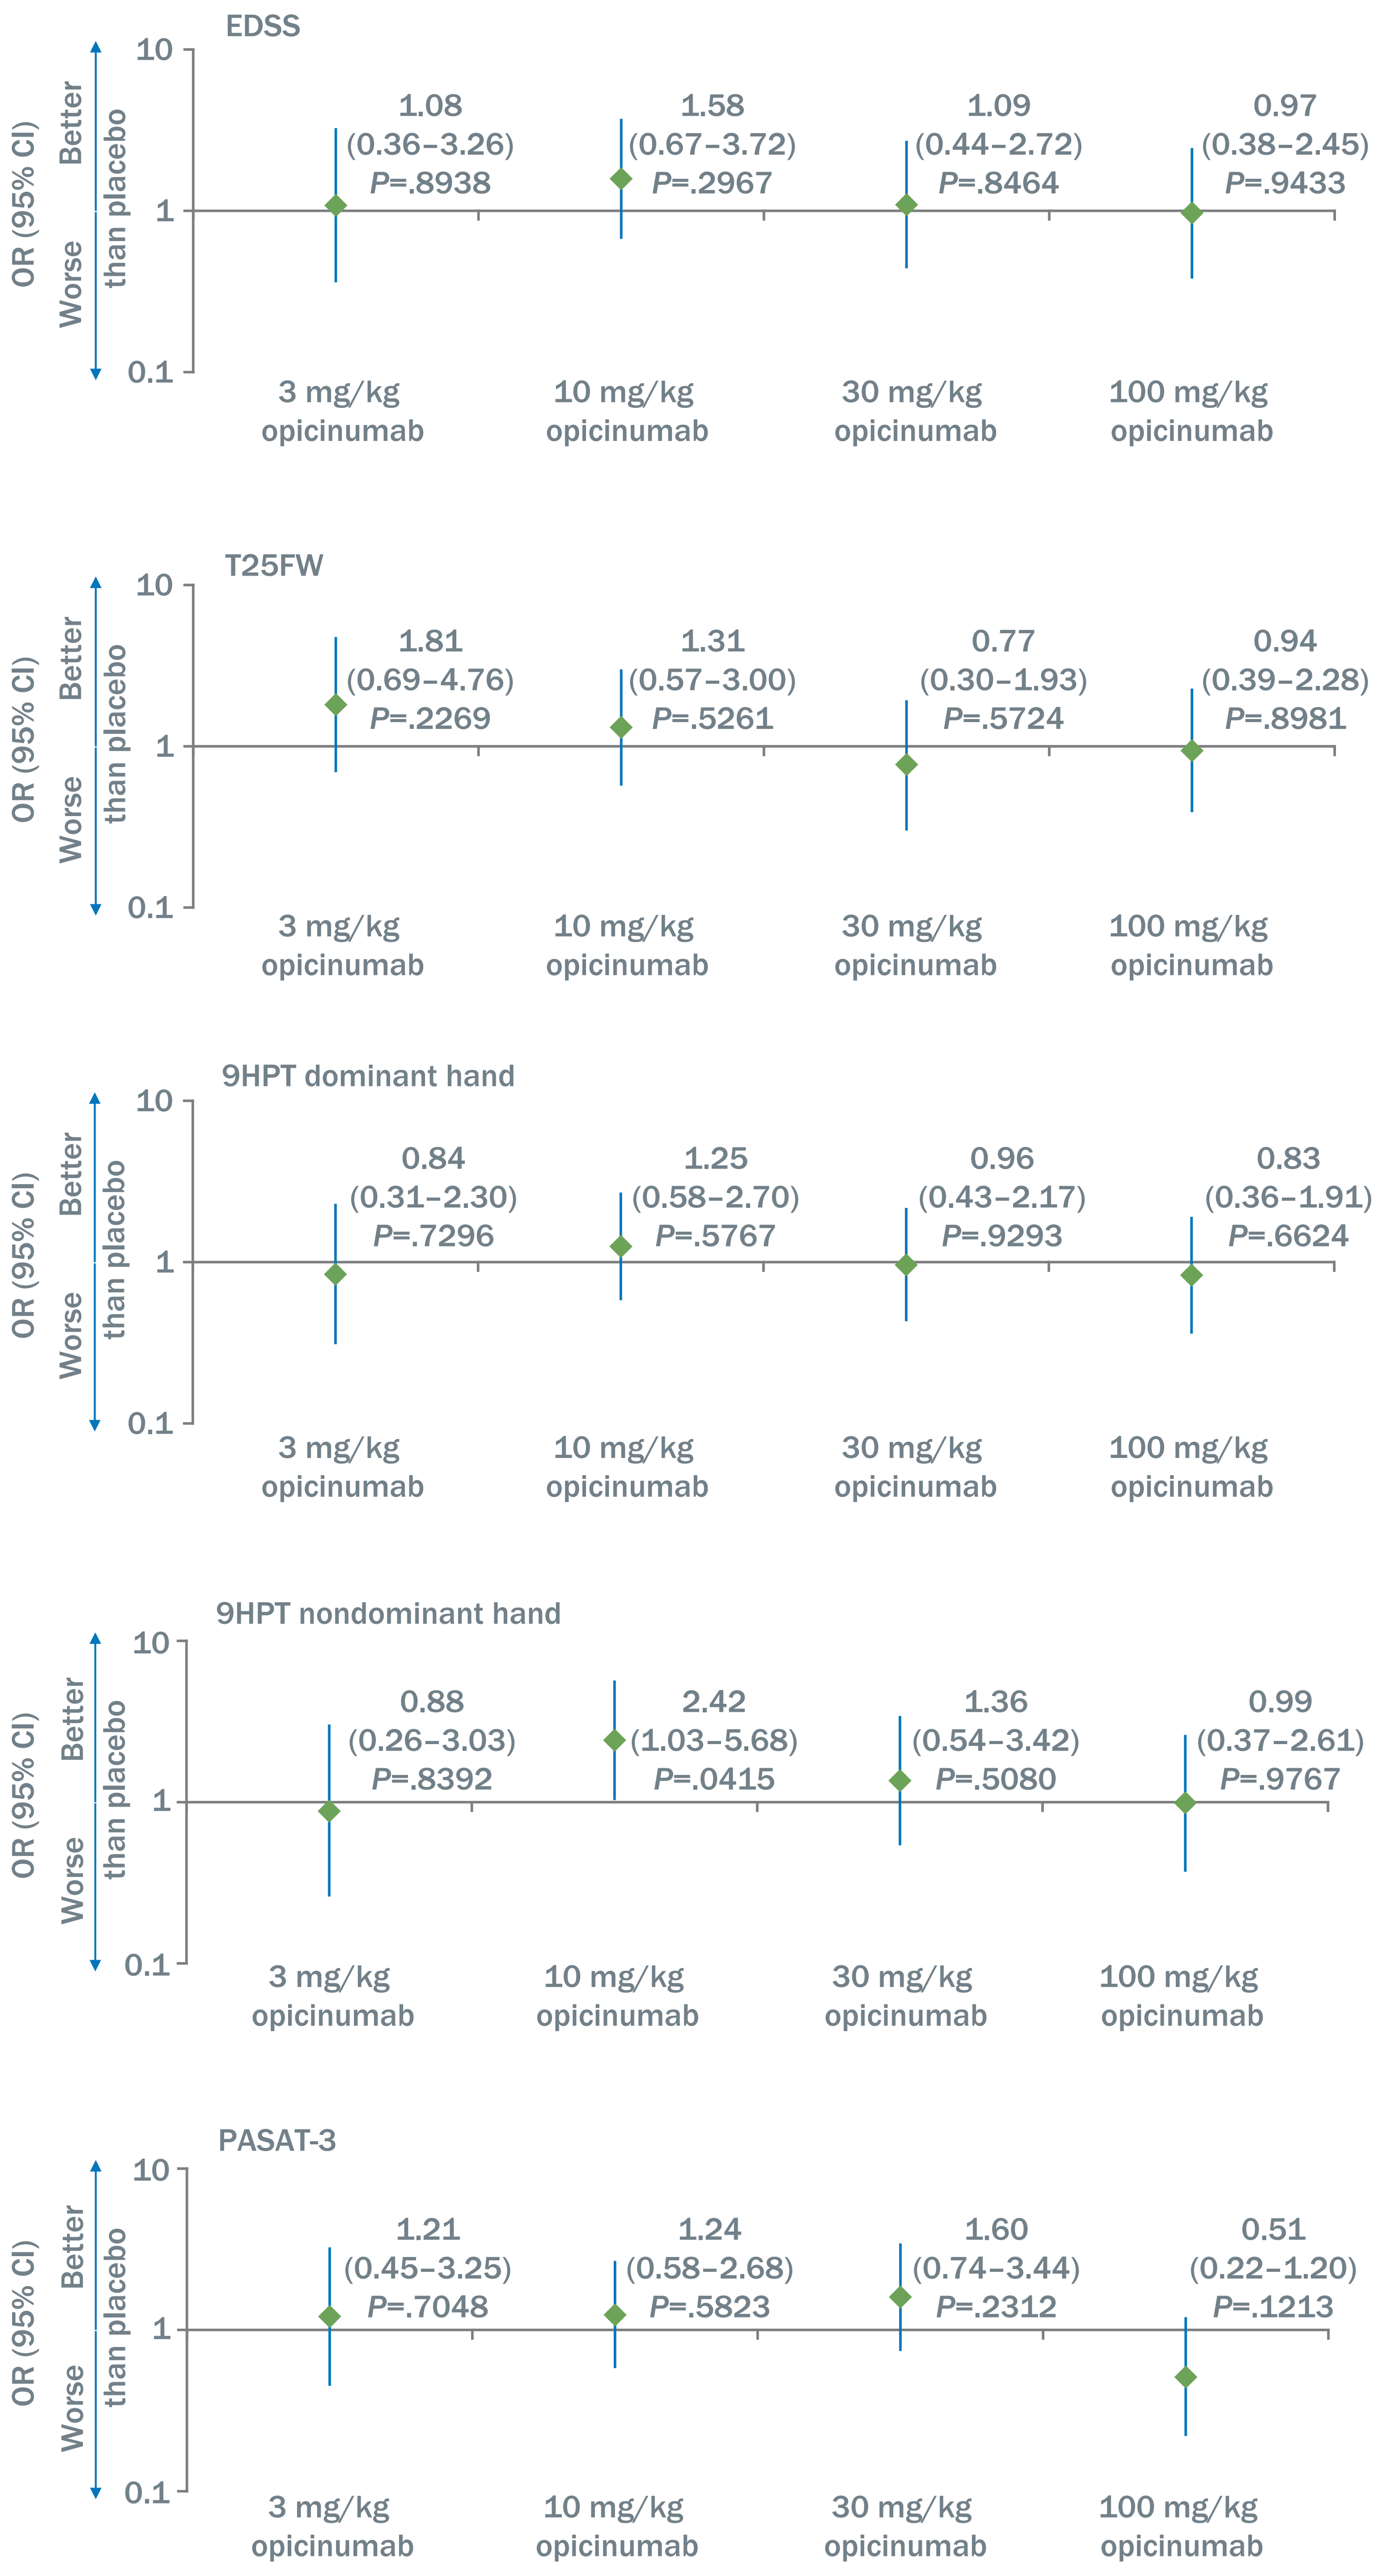
EDSS
OR (95% CI)
Better than placebo
Worse
10
1
0.1
1.08
(0.36–3.26)
P=.8938
1.58
(0.67–3.72)
P=.2967
1.09
(0.44–2.72)
P=.8464
0.97
(0.38–2.45)
P=.9433
3 mg/kg opicinumab
10 mg/kg opicinumab
30 mg/kg opicinumab
100 mg/kg opicinumab
T25FW
1.81
(0.69–4.76)
P=.2269
1.31
(0.57–3.00)
P=.5261
0.77
(0.30–1.93)
P=.5724
0.94
(0.39–2.28)
P=.8981
9HPT dominant hand
0.84
(0.31–2.30)
P=.7296
1.25
(0.58–2.70)
P=.5767
0.96
(0.43–2.17)
P=.9293
0.83
(0.36–1.91)
P=.6624
9HPT nondominant hand
0.88
(0.26–3.03)
P=.8392
2.42
(1.03–5.68)
P=.0415
1.36
(0.54–3.42)
P=.5080
0.99
(0.37–2.61)
P=.9767
PASAT-3
1.21
(0.45–3.25)
P=.7048
1.24
(0.58–2.68)
P=.5823
1.60
(0.74–3.44)
P=.2312
0.51
(0.22–1.20)
P=.1213

[a]Compared with placebo; based on a logistic regression model adjusted for MS type, region, and baseline component assessments

References 1. Cadavid D, *et al. Lancet Neurol*. 2017;16(3):189-199. 2. Tran JQ, *et al. Neuro Neuroimmunol Neuroinflammation*. 2014;1(2):e18. 3. Mellion M, *et al. Mult Scler*. 2015;23(suppl 11):P315.
Disclosures RH: advisor/speaker/consultant for Biogen, Merck Serono, Novartis, and Sanofi-Aventis; research support from Biogen and Merck Serono; KRE: consulting fees from and speaker bureaus for Biogen and Genzyme; research support from Biogen, Genentech, Novartis, Roche, and Sanofi-Genzyme; JD: advisor/speaker for Bayer HealthCare, Genzyme, Medis, Merck, Novartis, and Sanofi-Aventis; research support from the Ministry of Education and Science, Republic of Serbia (project no. 175031); XM: speaker fees/travel expense reimbursement from and steering committees/advisory boards for Actelion, Almirall, Bayer HealthCare, Biogen, Genzyme, Merck, Neuro-Tec, Novartis, Octapharma, Receptos, Roche, Sanofi-Aventis, Teva, and Trophos; H-PH: speaker fees/travel expense reimbursement from and steering committees/advisory boards for Biogen, GeNeuro, Genzyme, Merck, MedImmune, Novartis, Octapharma, Receptos, Roche, Sanofi-Aventis, and Teva; BB: advisory boards for and he/his institution has received honoraria from Biogen, Novartis, and Roche; research support to his institution from Actelion, Bayer HealthCare, Biogen, Genzyme, Novartis, and Roche; PAC: consulting fees from Biogen and Vertex; research support to his institution from Biogen, MedImmune, Novartis, and Teva; MM and DC: former employees of and hold stock in Biogen; this work is not related to DC's current employment with Fulcrum Therapeutics; RR, AI, YZ, and LX: employees of and hold stock/stock options in Biogen. **Acknowledgments** This study was sponsored by Biogen (Cambridge, MA, USA). Writing and editorial support for the preparation of this poster was provided by Excel Scientific Solutions (Horsham, UK): funding was provided by Biogen.

American Academy of Neurology 2017 - 69th Annual Meeting | April 22-28, 2017 | Boston, MA